AVA3996, a novel pre|CISION™ medicine, targets a warhead to the tumor microenvironment via Fibroblast Activation Protein-alpha (FAP-α) mediated cleavage to elicit tumor cell kill

David H Jones[1], Marine Houée[1], Hanna Buist[1], Sergi Marco[2], Chiara Braconi[2], Francis X Wilson[1], Fiona McLaughlin[1]
Avacta Therapeutics, London, United Kingdom[1]; University of Glasgow, Wolfson Wohl Cancer Research Centre, United Kingdom[2]

SCAN

Introduction

- AVA3996 is a therapeutic entity based on Avacta's proprietary pre|CISION™ technology which incorporates a substrate that is sensitive to cleavage by FAP-α.
- FAP-α, a post-prolyl endopeptidase, is overexpressed on the surface of activated fibroblastic cells which are abundant in the supporting stroma of over 90% of malignant epithelial cancers.
- The pre|CISION™ substrate can be utilized in a drug conjugate linker or to generate masked warheads that are only activated in the tumor microenvironment.
- AVA3996 consists of a proteasome inhibitor molecule (an analog of Bortezomib) covalently bonded to a peptide containing a cleaving sequence (D-Ala-L-Pro), which is designed to be susceptible to hydrolysis by Fibroblast Activation Protein α (FAP-α) but is resistant to hydrolysis by both closely related and wider mammalian peptidases.
- Proteasome inhibitors are a first line of treatment for certain hematologic indications such as multiple myeloma. However, clinical utility of proteasome inhibitors is limited by severe dose-limiting toxicities, including peripheral neuropathy.
- AVA3996 has the potential to deliver elevated, effective levels of proteasome inhibitor directly to the solid tumor microenvironment while reducing systemic exposure and hence associated toxicities.

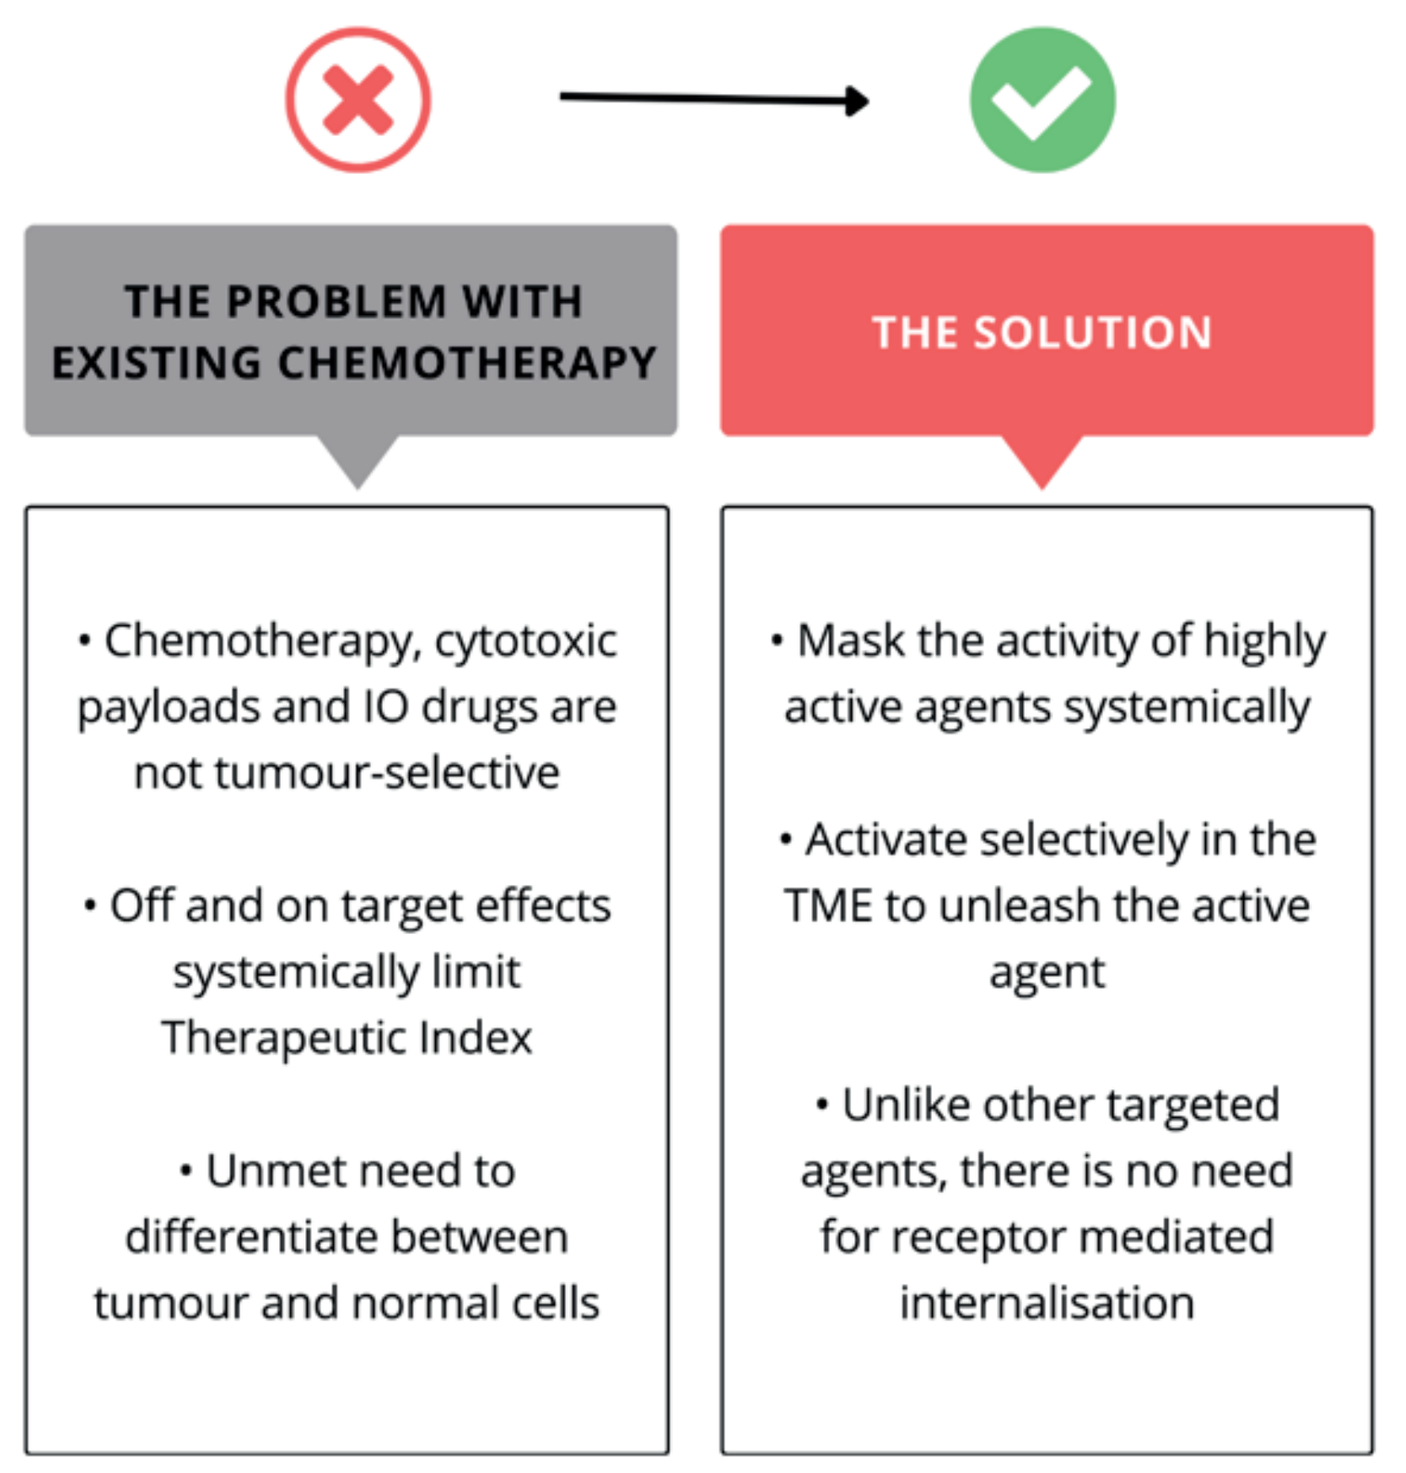
THE PROBLEM WITH EXISTING CHEMOTHERAPY
• Chemotherapy, cytotoxic payloads and IO drugs are not tumour-selective
• Off and on target effects systemically limit Therapeutic Index
• Unmet need to differentiate between tumour and normal cells
THE SOLUTION
• Mask the activity of highly active agents systemically
• Activate selectively in the TME to unleash the active agent
• Unlike other targeted agents, there is no need for receptor mediated internalisation

Cellular proteasome activity is inhibited after AVA3996 cleavage, resulting in cell death

- Proteasome inhibition may be measured in a lysed-cell assay.
- AVA3996 exhibits a low level of activity in cells not expressing FAP-α, but may be cleaved by exogenous FAP-α resulting in release of AVA2727D and subsequent proteasome inhibition.

Figure 1. Following cell lysis, proteasome activity is measured by cleavage of an AMC-tagged peptide substrate. HEK cells are either transfected with a FAP-α expressing plasmid or with empty vector.

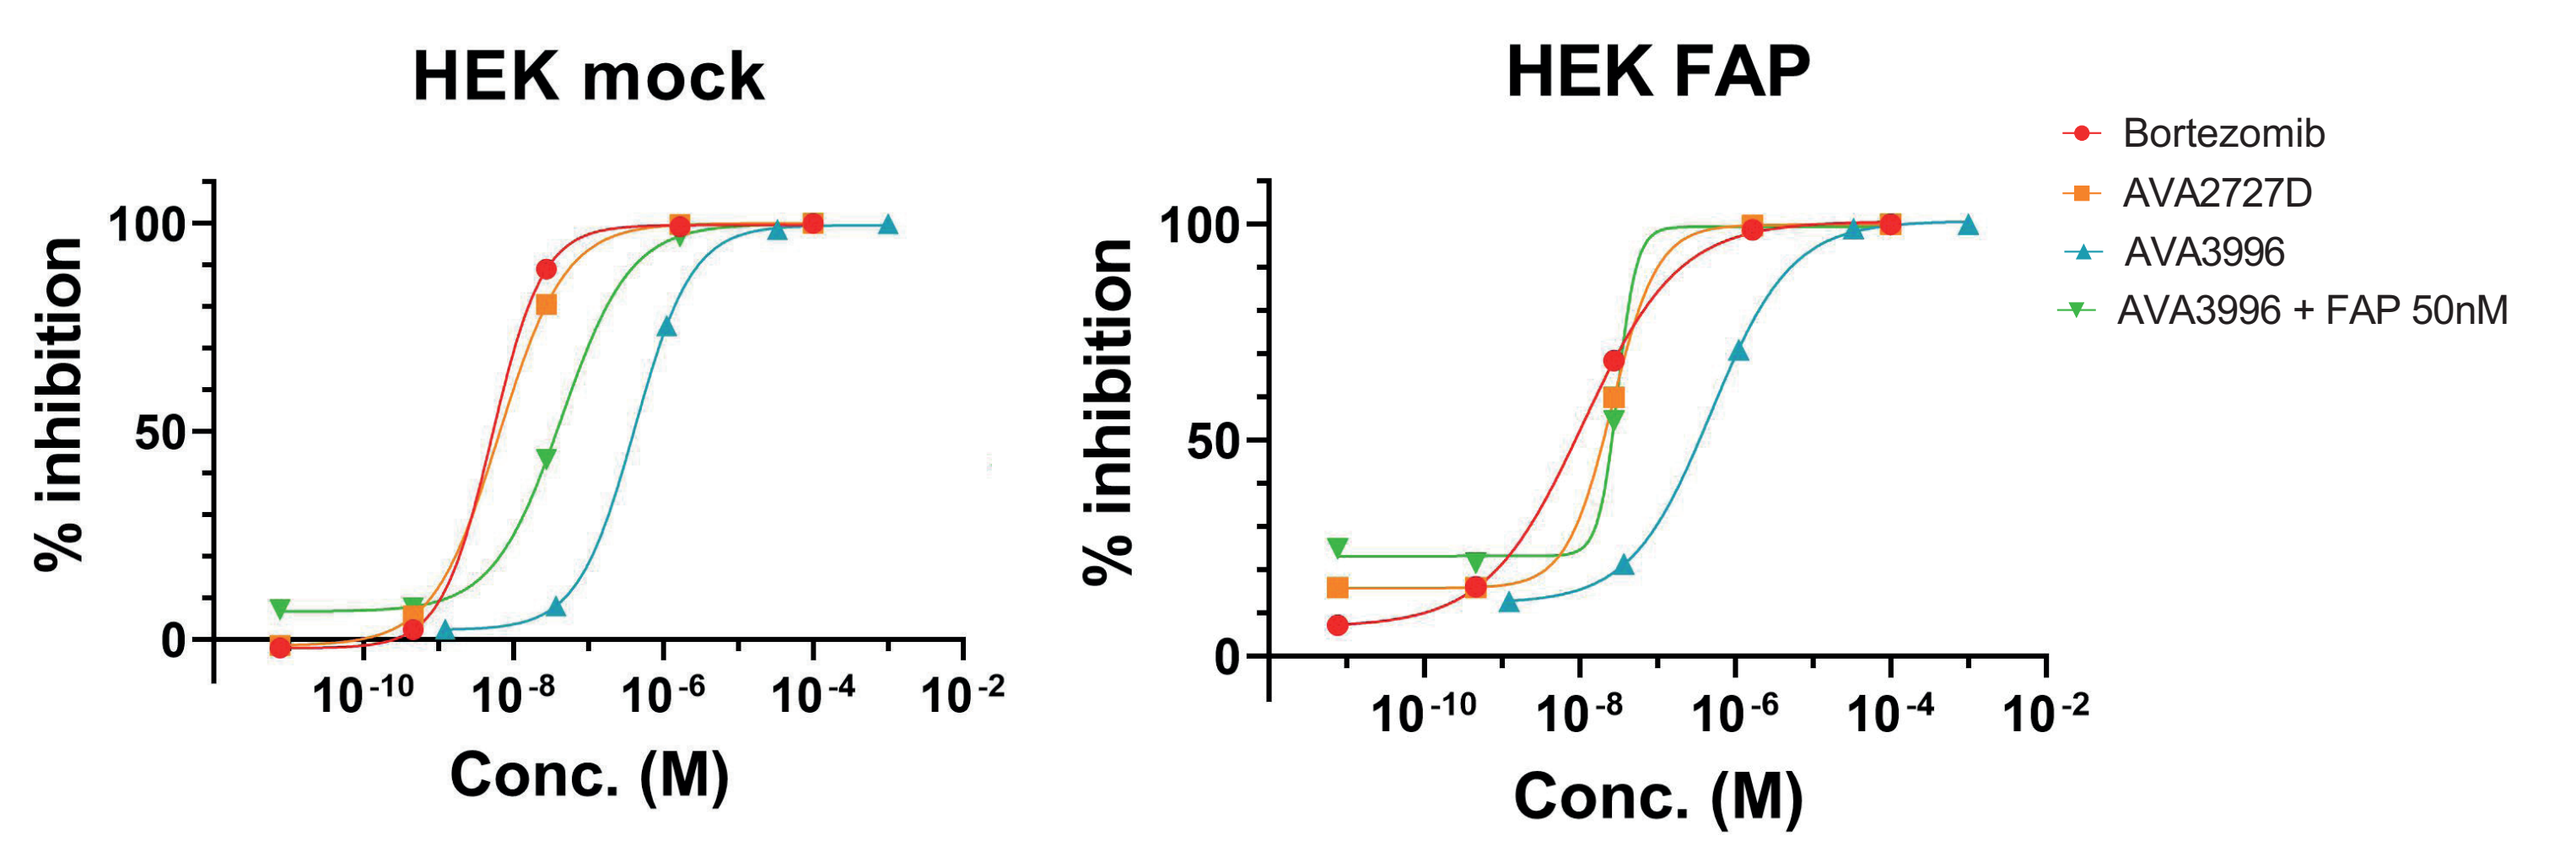
HEK mock
HEK FAP
% inhibition
Conc. (M)
Bortezomib
AVA2727D
AVA3996
AVA3996 + FAP 50nM

- Cell viability was measured in a Cell Titer Glo assay.
- In cells which do not express FAP-α, AVA3996 again shows a low level of activity.
- Upon incubation with exogenous FAP-α, the cytotoxicity of AVA3996 increases ~100-fold to show equivalent activity to the active warhead AVA2727D.
- HEK cells which over-express FAP-α (HEK hFAP) are also able to cleave AVA3996 in the absence of exogenous FAP-α and hence exhibit sensitivity equivalent to AVA2727D.

EC_{50} (nM)	Bortezomib	AVA2727D	AVA3996	AVA3996 + FAP	AVA3996 + FAPi
HEK mock	4.29	34.72	3027	19.2	3482
HEK FAP	4.22	22.75	47.1	14.4	1190

Figure 2 and Table 1. Cell viability (Cell Titer Glo assay) following compound exposure for 48 hours in serum-free conditions.

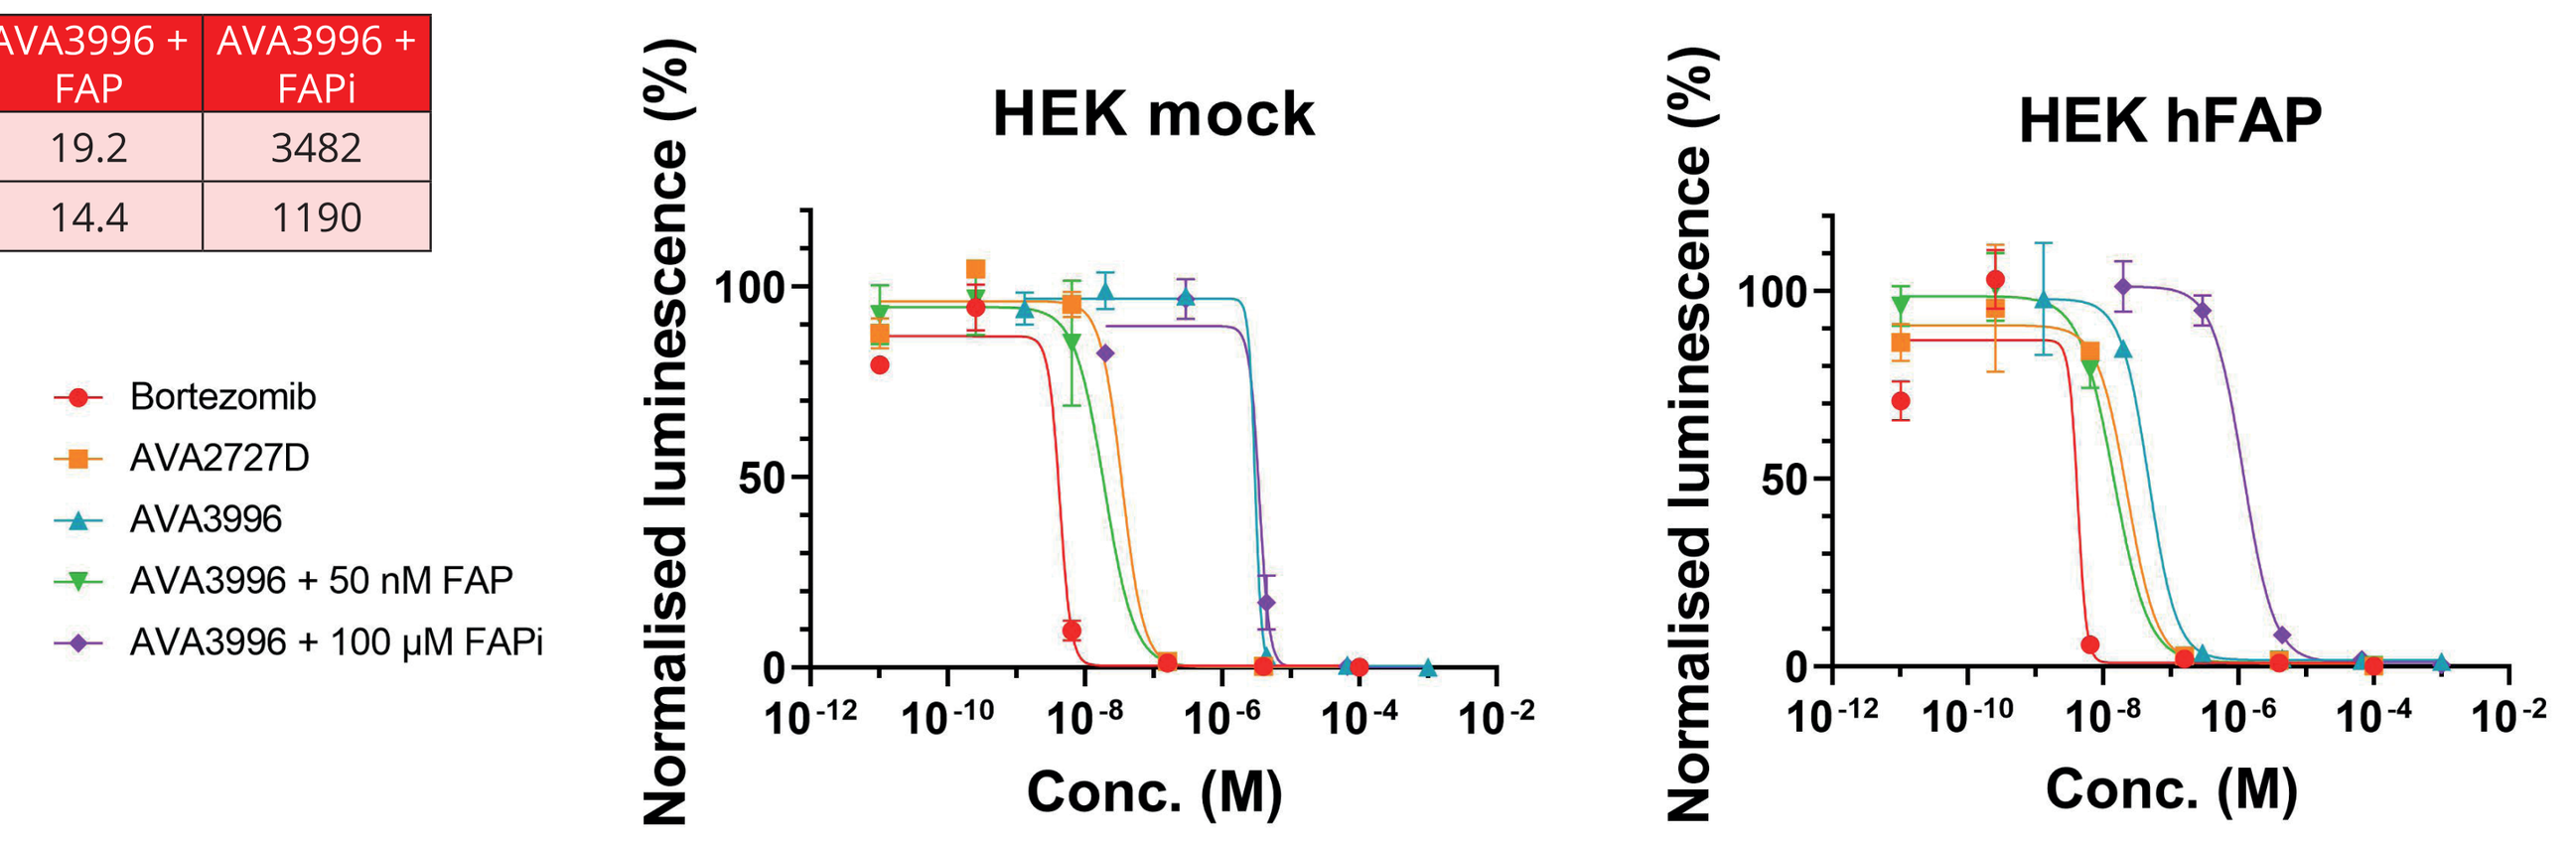
Bortezomib
AVA2727D
AVA3996
AVA3996 + 50 nM FAP
AVA3996 + 100 µM FAPi
HEK mock
HEK hFAP
Normalised luminescence (%)
Conc. (M)

FAP mediated hydrolysis of AVA3996 releases free AVA2727D proteasome inhibitor (PI)

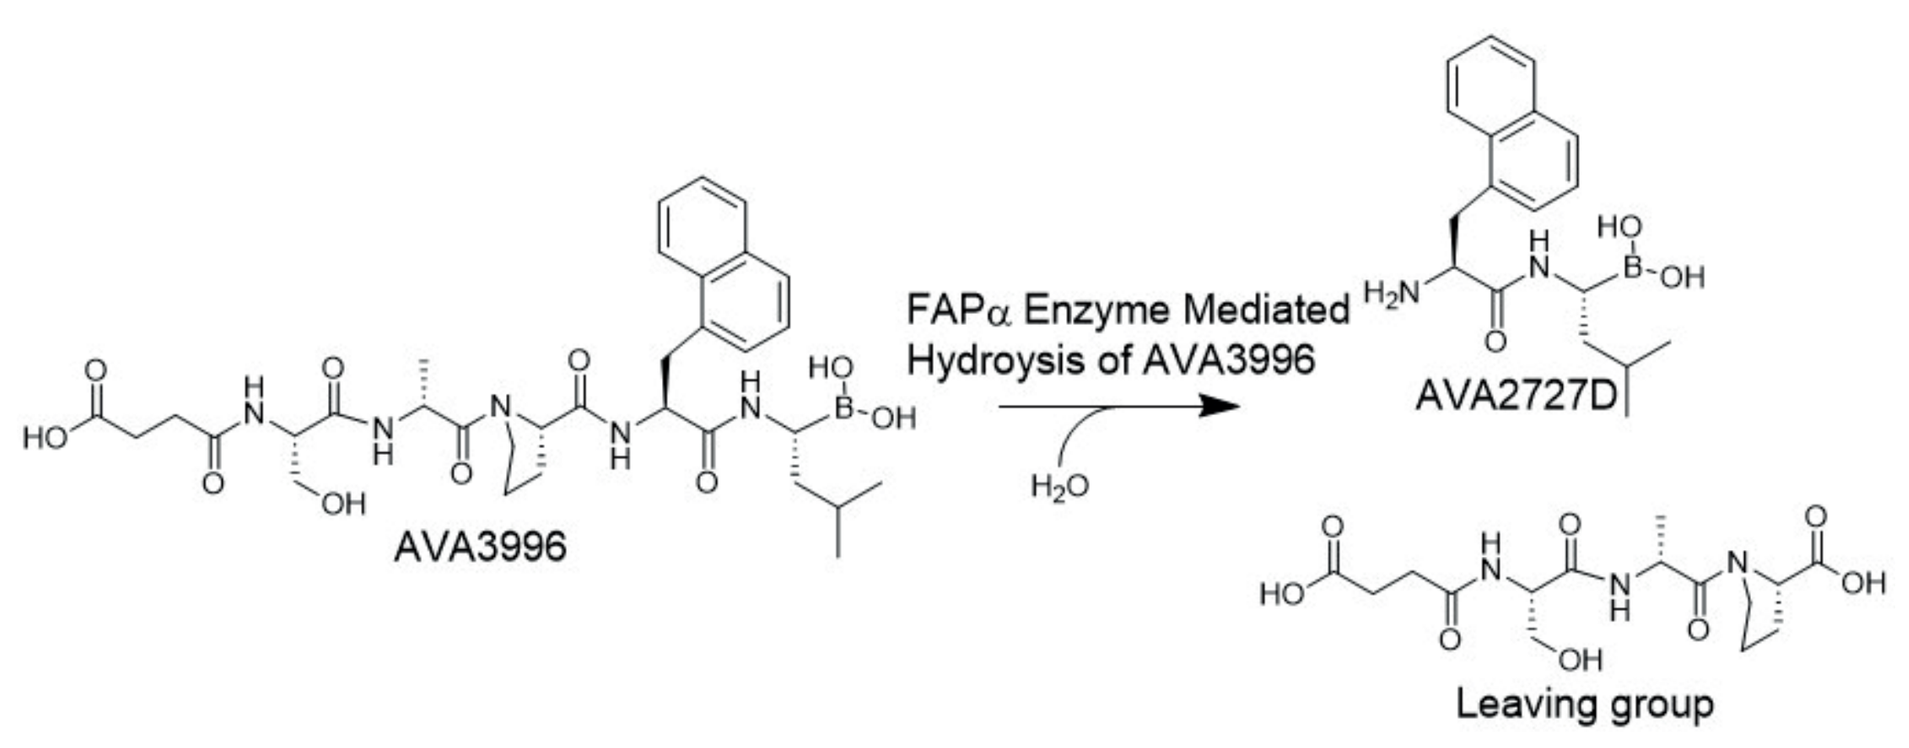
AVA3996
FAPα Enzyme Mediated Hydroysis of AVA3996
AVA2727D
Leaving group

Figure 3a. Structures of AVA3996 and the active warhead AVA2727D which is generated following cleavage by FAP-α.

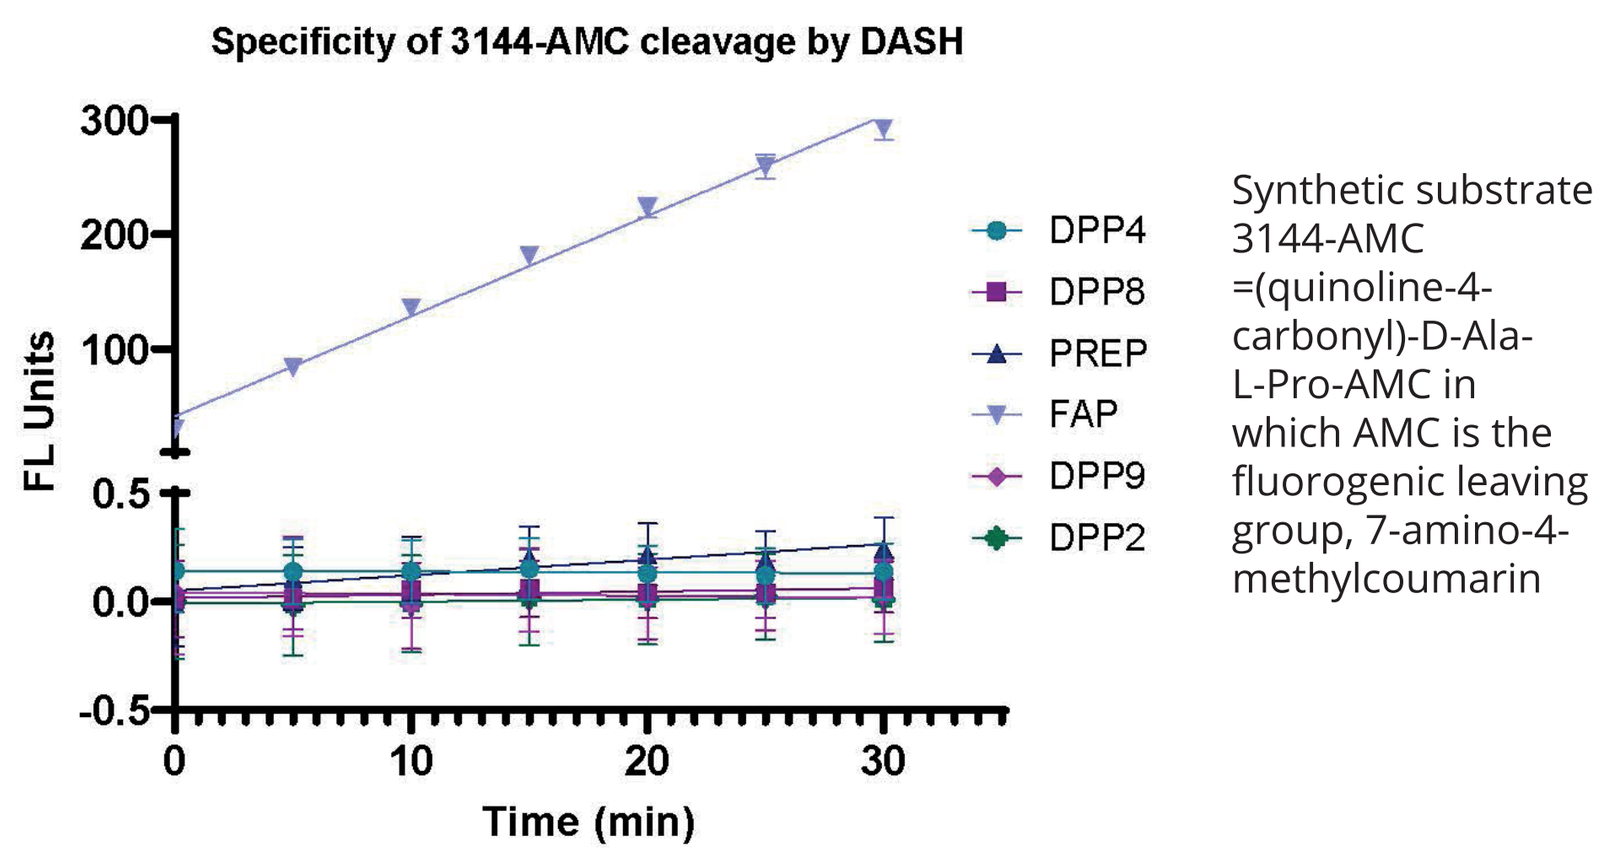
Specificity of 3144-AMC cleavage by DASH
FL Units
Time (min)
DPP4
DPP8
PREP
FAP
DPP9
DPP2
Synthetic substrate 3144-AMC =(quinoline-4-carbonyl)-D-Ala-L-Pro-AMC in which AMC is the fluorogenic leaving group, 7-amino-4-methylcoumarin

Figure 3b: The linker is exquisitely selective for cleavage by the protease FAP-α and not by closely related proteases.

AVA3996 shows greater cell cytotoxicity when cancer cells are co-cultured with fibroblasts

- Stromal regions of cholangiocarcinoma (CCA) typically express high levels of FAP-α.
- Co-culture was established with four different CCA cell lines (two shown below) and immortalised cancer-associated fibroblasts (CAFs). Conditions were established for serum-free growth in the presence of media supplements. FAP-α in this system is provided by endogenous levels on the CAFs.
- CCA cells are more sensitive to warhead AVA2727D than to AVA3996 in monoculture.
- AVA3996 exhibits enhanced cell kill in the presence of CAFs which provide FAP-α; there is no impact on AVA2727D activity in co-culture.

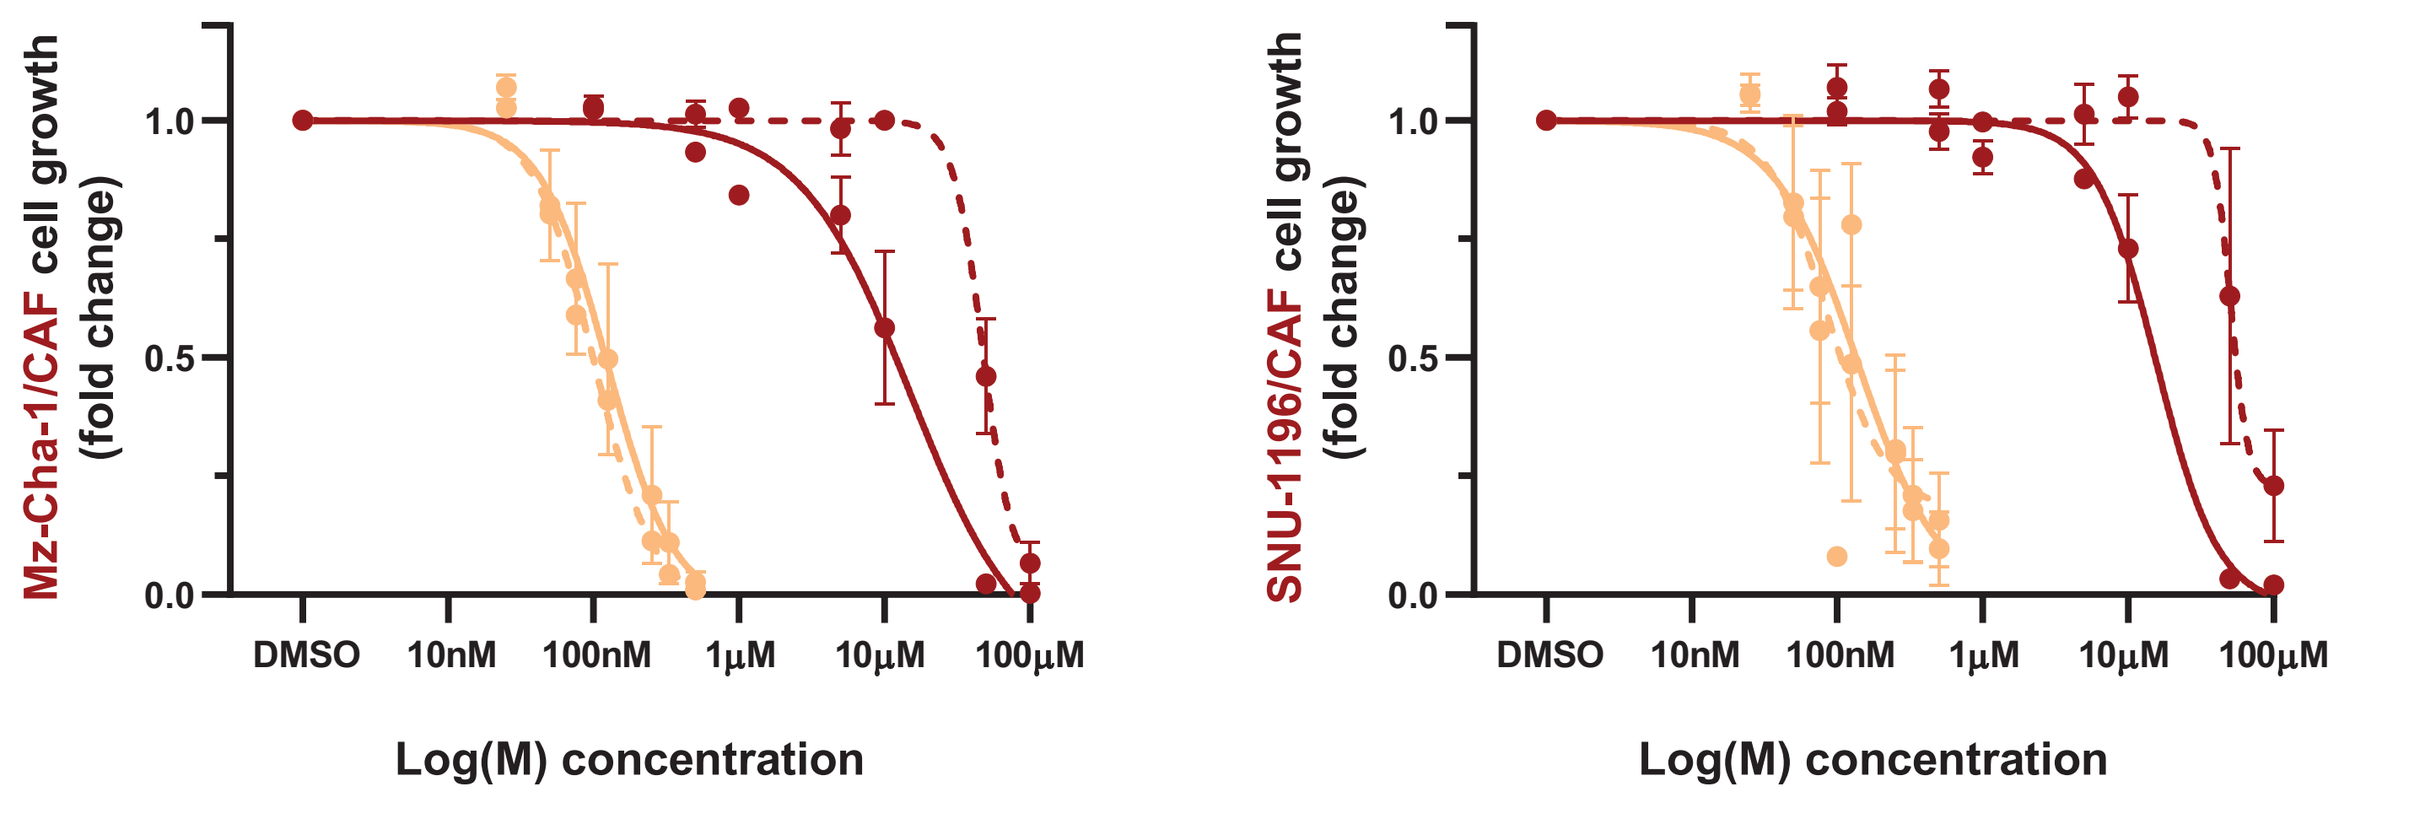
Mz-Cha-1/CAF cell growth (fold change)
SNU-1196/CAF cell growth (fold change)
DMSO
10nM
100nM
1µM
10µM
100µM
Log(M) concentration
AVA3996 coculture
AVA3996 monoculture
AVA2727D coculture
AVA2727D monoculture

Figure 4. Sensitivity of CCA cells in mono-culture and co-culture with fibroblasts when exposed to AVA3996 and AVA2727D.

AVA3996 shows efficacy in high FAP *in vivo* PDX models with no toxicity (as observed with Bortezomib)

- Patient-derived xenograft models were selected based on high FAP-α expression.
- Initial studies showed efficacy in three models: melanoma, rhabdomyosarcoma and colorectal (Figure 5).
- In some models (including the melanoma model), Bortezomib showed toxicity as evidenced by decreased body weight. This was not seen for AVA3996.

Figure 5. Tumor growth inhibition upon administration of AVA3996. AVA3996 dosed sc BIW for 6 doses.

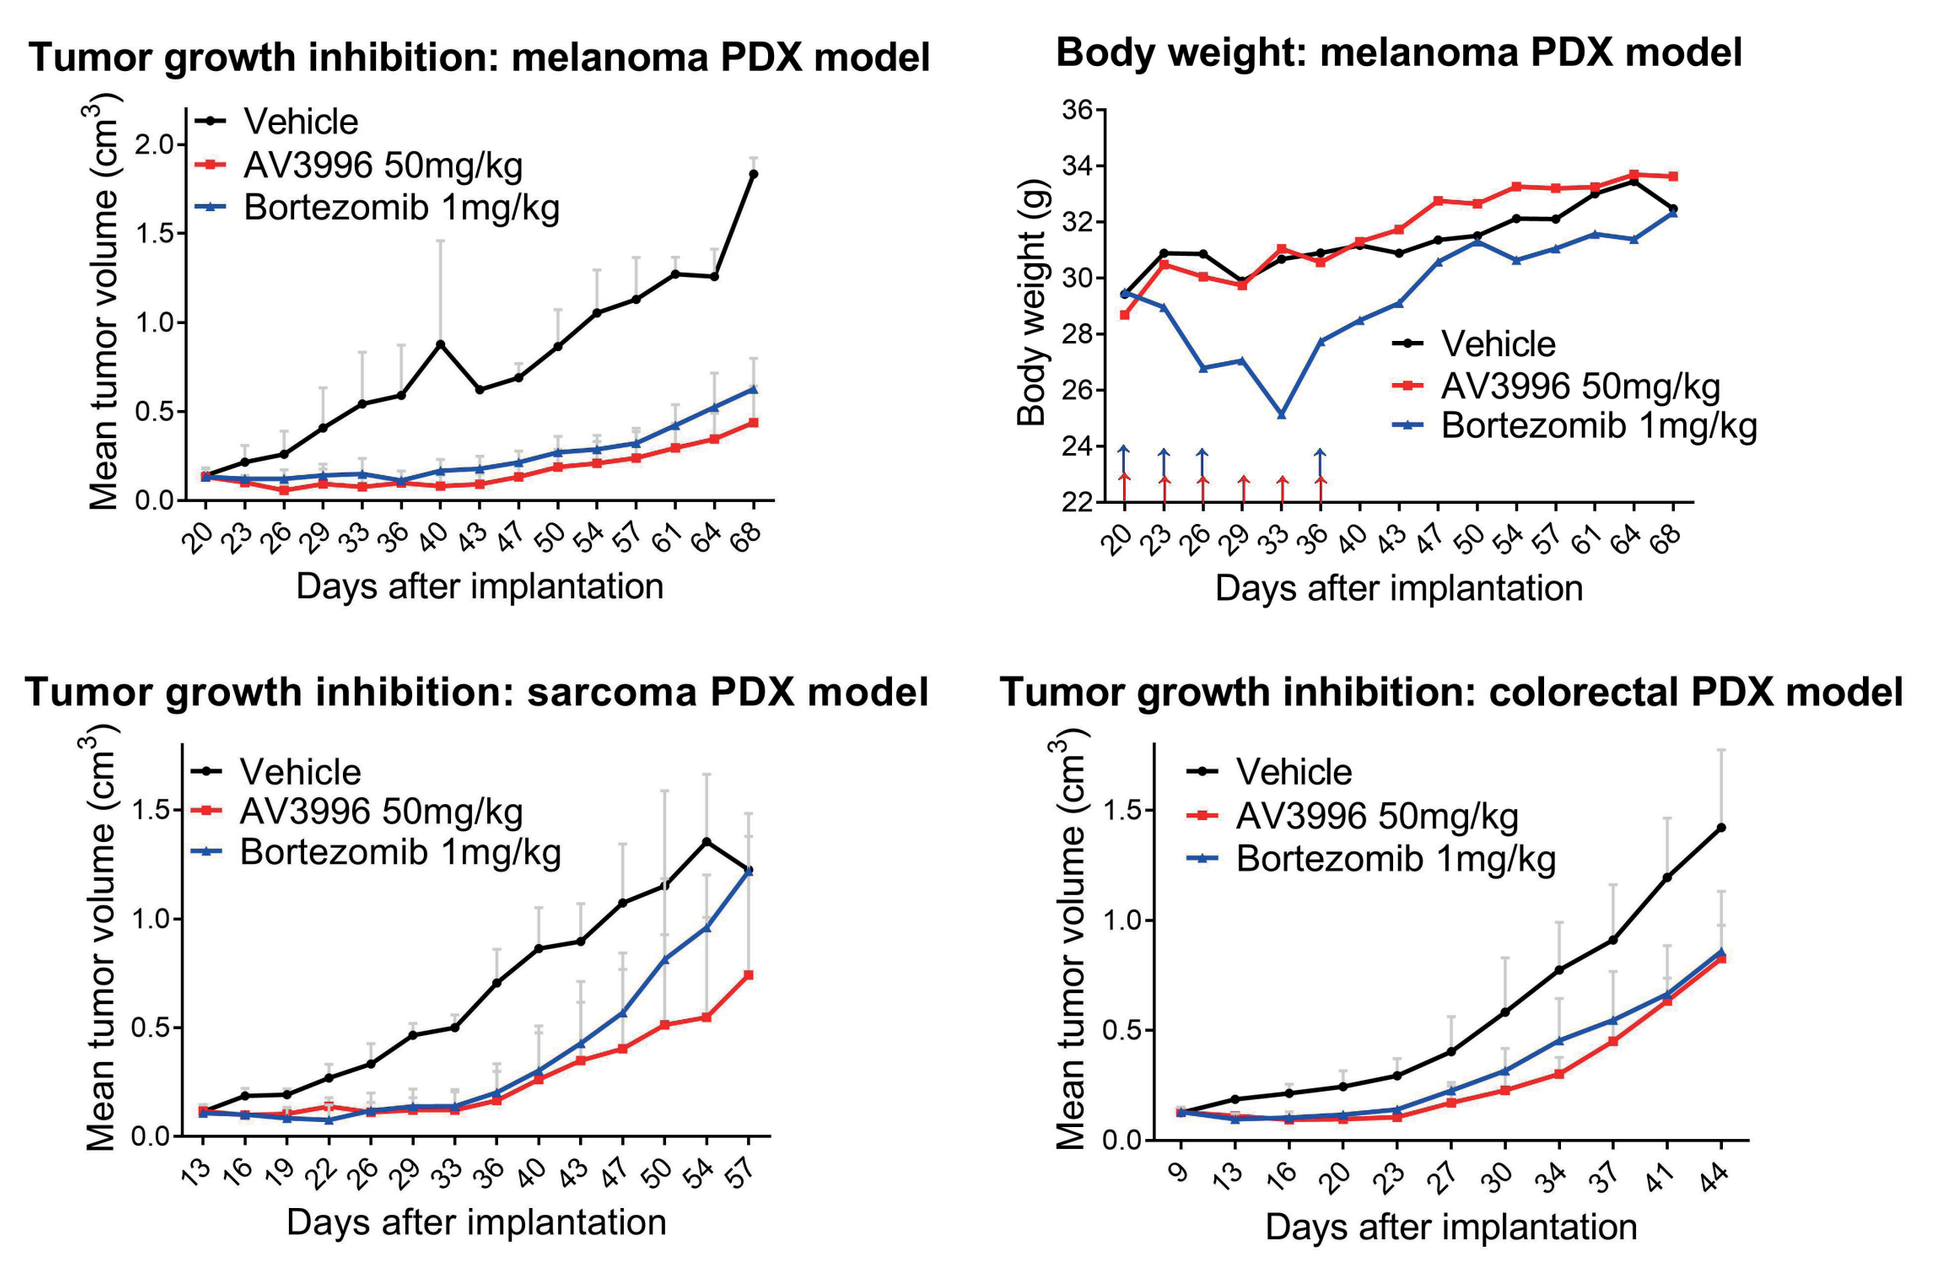
Tumor growth inhibition: melanoma PDX model
Body weight: melanoma PDX model
Tumor growth inhibition: sarcoma PDX model
Tumor growth inhibition: colorectal PDX model
Vehicle
AV3996 50mg/kg
Bortezomib 1mg/kg
Mean tumor volume (cm³)
Body weight (g)
Days after implantation

AVA3996 is selectively cleaved at the tumor resulting in tumor growth inhibition in a melanoma PDX model

- A follow-up study was performed in the V600E BRAF mutant melanoma model. AVA3996 showed similar tumor growth inhibition to Trametinib.
- In a satellite group, blood samples were taken at 30min, 1hr and 4hrs, and tumors at 4hrs.
- AVA3996 is rapidly cleared from the blood while AVA2727D is only observed at very low levels.
- In the tumor, cleavage of AVA3996 by FAP-α results in high levels of warhead AVA2727D, hence cytotoxicity.

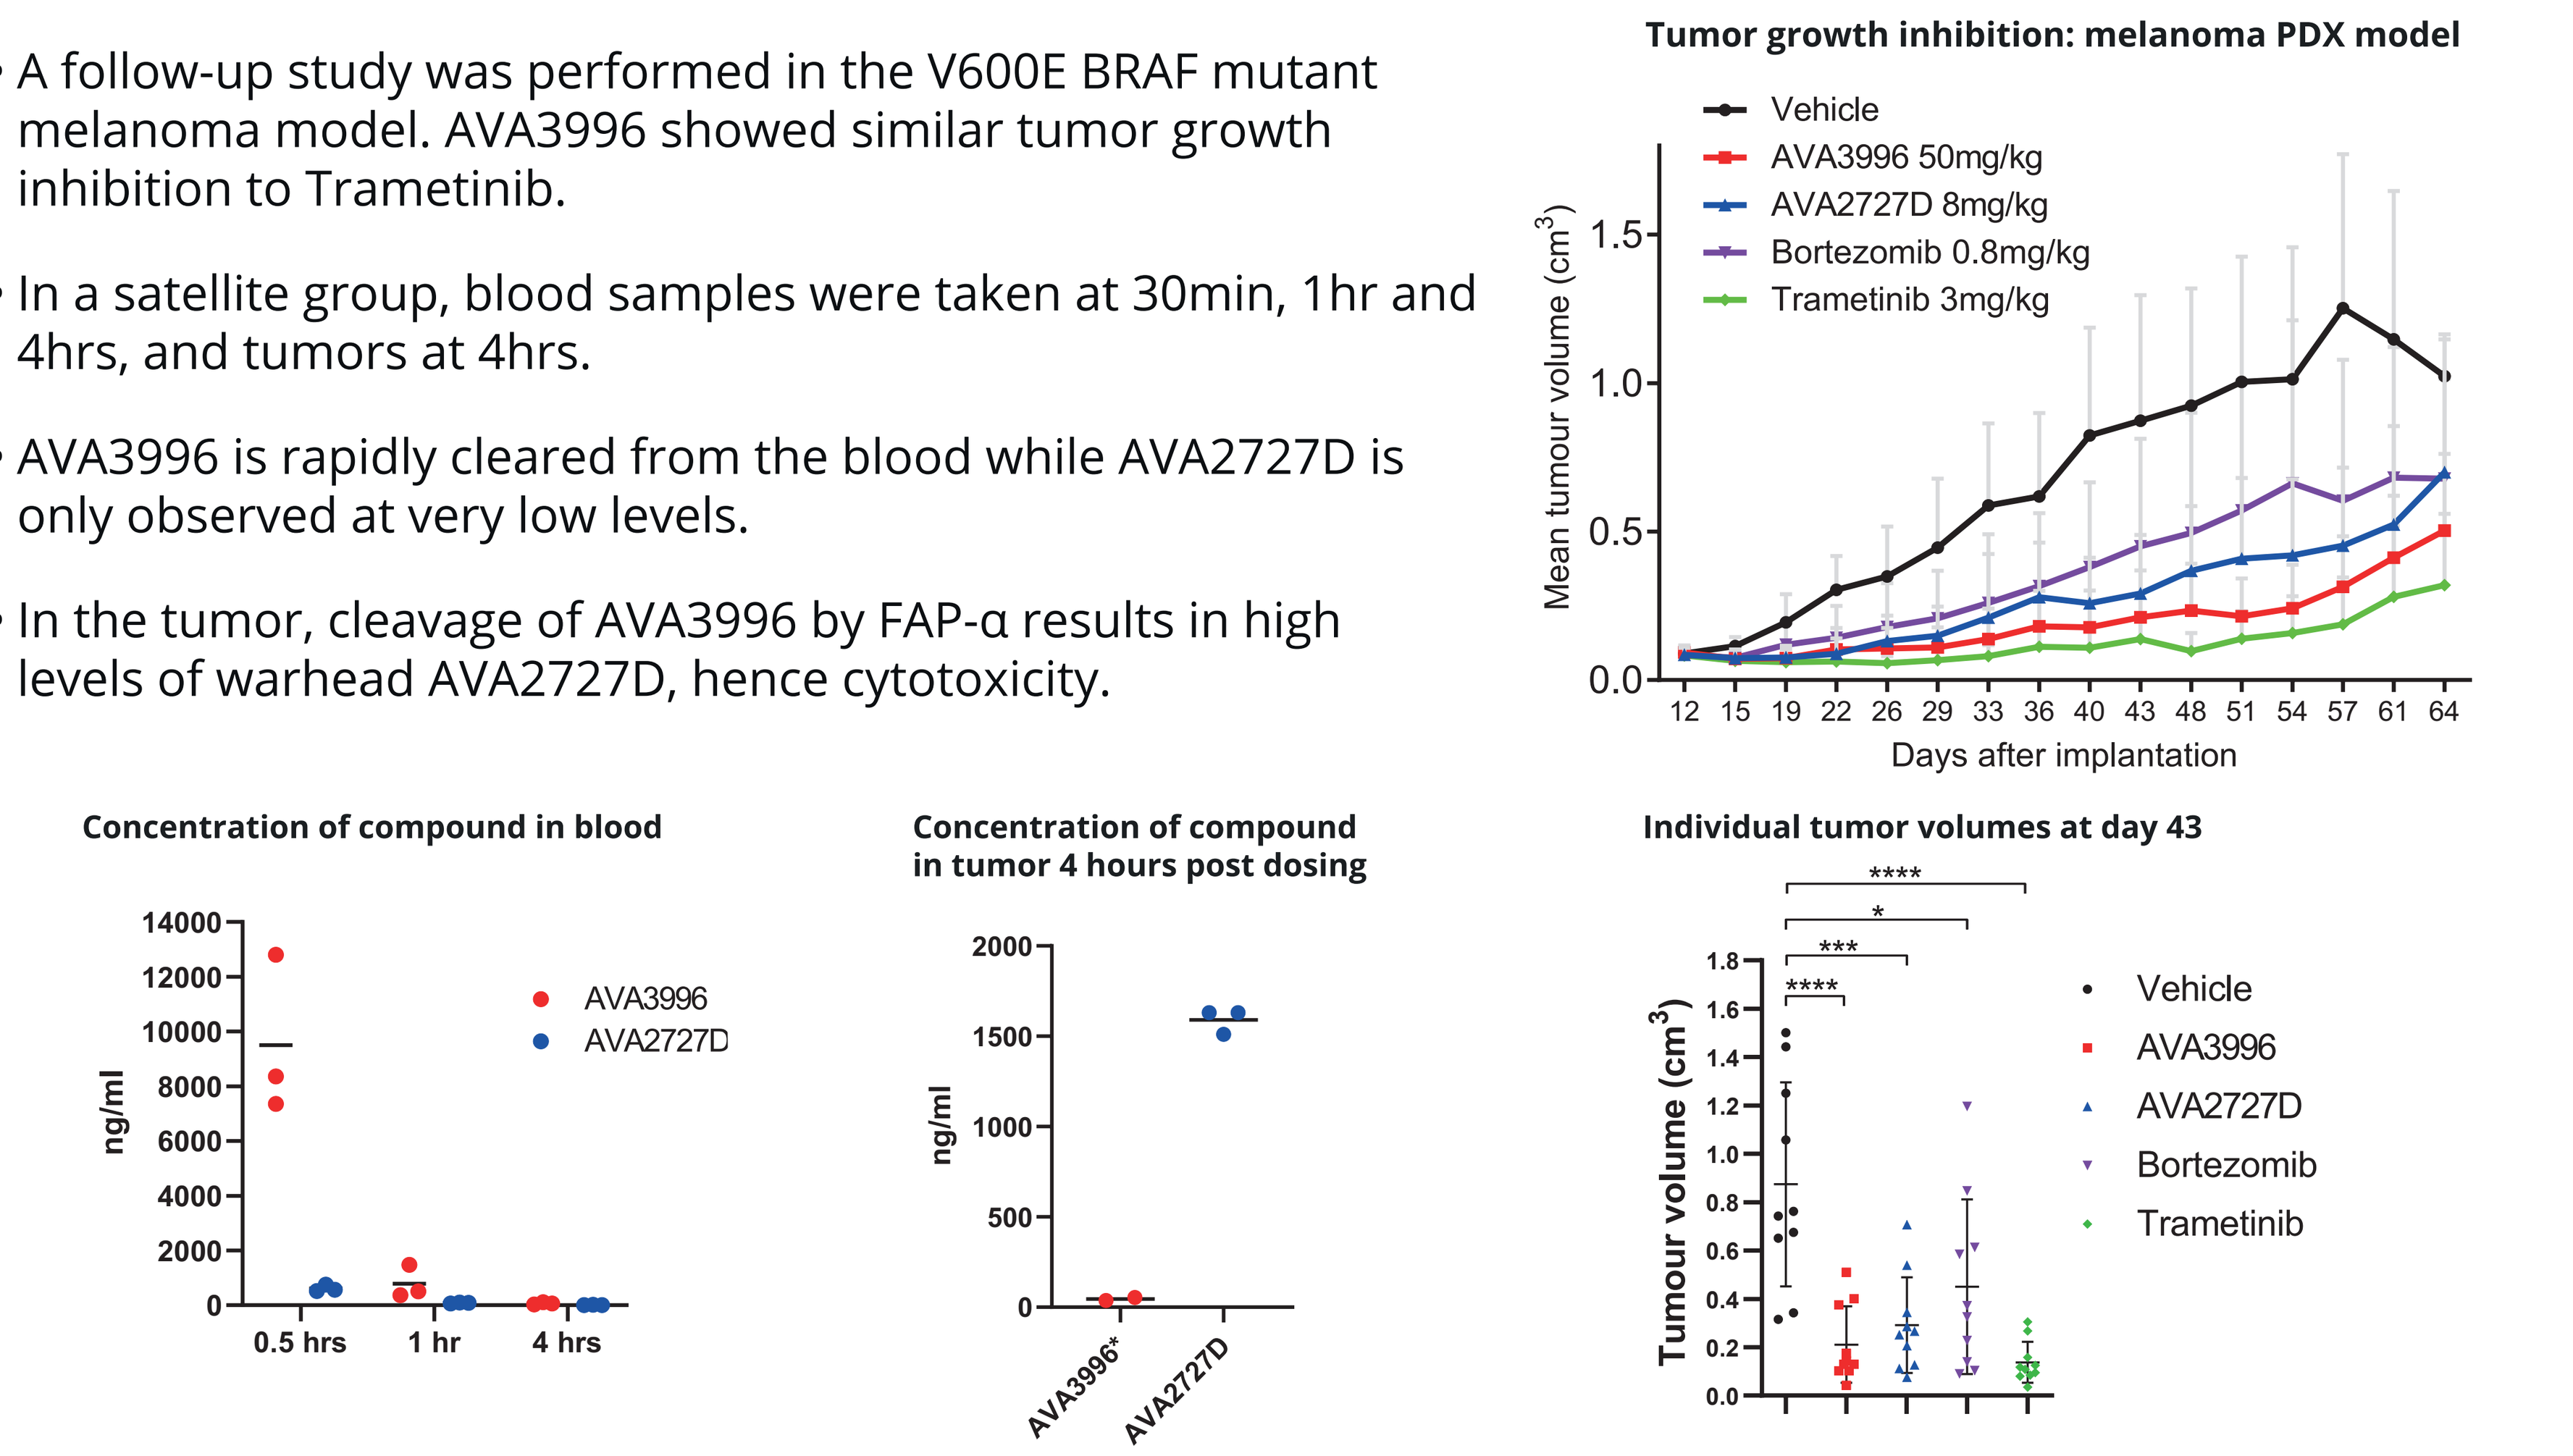
Tumor growth inhibition: melanoma PDX model
Vehicle
AVA3996 50mg/kg
AVA2727D 8mg/kg
Bortezomib 0.8mg/kg
Trametinib 3mg/kg
Mean tumour volume (cm³)
Days after implantation
Concentration of compound in blood
ng/ml
0.5 hrs
1 hr
4 hrs
AVA3996
AVA2727D
Concentration of compound in tumor 4 hours post dosing
AVA3996*
AVA2727D
Individual tumor volumes at day 43
Tumour volume (cm³)
Vehicle
AVA3996
AVA2727D
Bortezomib
Trametinib

Figure 6. Tumor growth inhibition in a melanoma model with analysis at day 43 (last day when all vehicle mice were still on study); concentrations of AVA3996 and AVA2727D in blood and tumor.

IND-enabling studies

- In CEREP safety screen, AVA3996 and AVA2727D showed no activity against all targets.
- The Table below shows TK parameters following intravenous administration in rats.
- Rat MTD: AVA3996 MTD 37.5mg/kg and was well tolerated at 25mg/kg; AVA2727D MTD 4mg/kg.
- Dog toxicity: AVA3996 was well tolerated at 3.75mg/kg.

Dose of AVA3996 administered (mg/kg) [equivalent AVA2727D content]	AVA3996		AVA2727D	
	Cmax (ng/ml)	AUCtlast (ng.h/ml)	Cmax (ng/ml)	AUCtlast (ng.h/ml)
8.3 [4]	18350	2770	117	84
25 [12]	78825	10975	258	237
37.5 [18]	123150	19625	367	394

Summary

- AVA3996 is cleaved by FAP-α with high specificity: no safety issues are apparent with AVA3996 or the active warhead AVA2727D.
- AVA3996 is activated when exposed to cells expressing FAP-α or when exposed to cells in the presence of fibroblasts (which express FAP-α). Activation by FAP-α elicits tumor cell death.
- Cleavage of AVA3996 by FAP-α is observed in tumors in a PDX model. In this model, AVA3996 exhibits tumor growth inhibition without the toxicity observed for Bortezomib.
- AVA6000 is the most advanced therapeutic based on Avacta's pre|CISION™ technology, currently in Phase I clinical trials. Further CMC, safety, efficacy and disease positioning studies are ongoing for AVA3996 with the aim to advance this to the clinic.

POS039 For further information please visit www.avacta.com Poster ID: C169